In vivo engineering of CAR T cells using a novel targeted LNP-mRNA technology

Gregor Adams, Ferran Soldevila, Daiki Matsuda, Yan Zhang, Yanjie Bao, Brittany Ross, Stuart A. Sievers, John Li, Michael Peel, Michelle Nguyen, Barzan A. Sadiq, Josephine Nguyen, Theresa Hunter, Claudia Fernandez, Yi Kuo, James Vestal, Matthew Butcher, Jeffrey Chen, Stanley Zhang, Duy Nguyen, Makan Khoshnejad, Donald Jhung, Steve Tanis, Michael Rosenzweig, Priya Karmali, Adrian Bot, Haig Aghajanian
Capstan Therapeutics, San Diego, CA

BACKGROUND

Autologous chimeric antigen receptor (CAR) T cell therapies have revolutionized the treatment of some cancers and are now demonstrating effects in autoimmune disease. With several approved therapies on the market and cure rates approaching 50% in various hematologic malignancies, access to these life-saving therapies is paramount. However, need for lymphodepletion conditioning, narrow access, and clinical performance limit broader applicability of ex vivo engineered CAR T cell products. Utilizing the success of mRNA lipid nanoparticles (LNP) as COVID vaccines, Capstan Therapeutics has developed a non-viral novel targeted LNP (tLNP) platform, that is purpose-built for specific delivery of therapeutic CAR in mRNA format to immune cells through functionalization with a targeting antibody. Through access, scalability and clinical performance, in vivo CAR products may enable much broader applicability of this therapeutic modality, including earlier lines of treatment and use in non-oncologic indications.

CAPSTAN TARGETED LIPID NANOPARTICLE (tLNP)

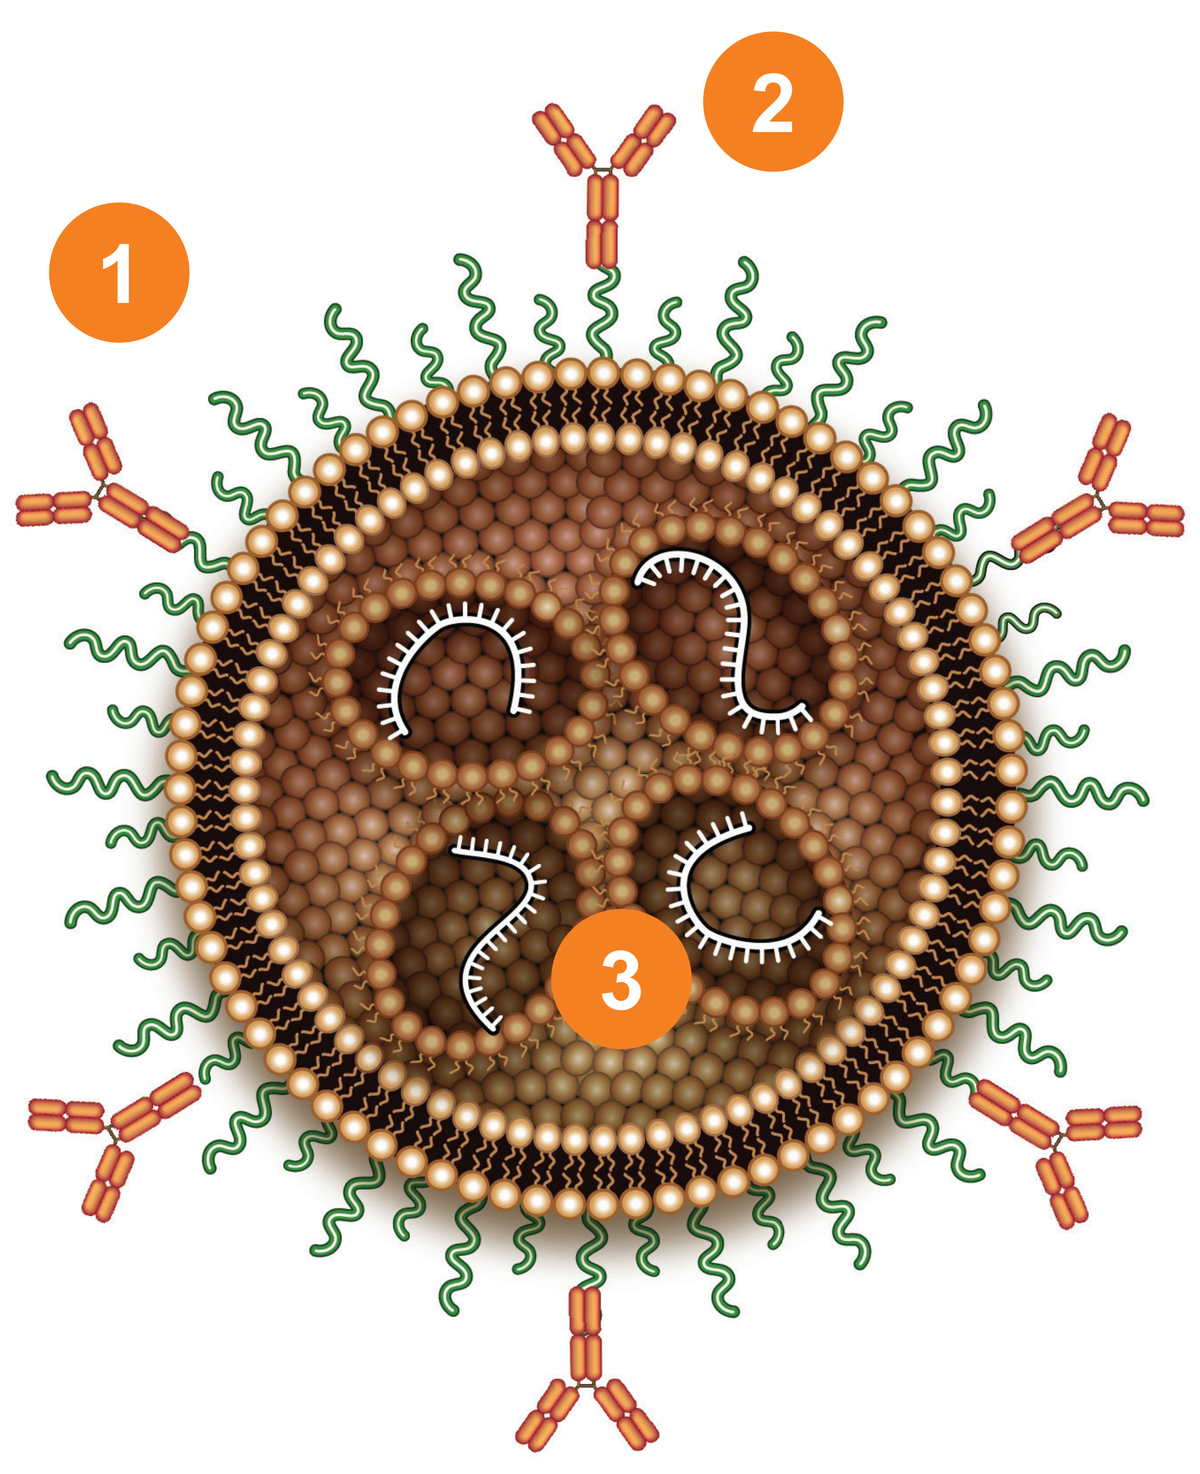
1
2
3

OUR tLNP PLATFORM IS MADE UP OF THREE COMPONENTS:

1. LNP DELIVERY VEHICLE

Capstan's proprietary lipid nanoparticle is a non-viral system designed for increased tolerability and biodegradability to allow for repeat in vivo dosing.

2. CELL TYPE-SPECIFIC TARGETING BINDERS

Antibody or antibody fragments functionalized onto the nanoparticle surface, creating targeted lipid nanoparticles (tLNPs) for efficient delivery of disease-specific payloads.

3. DISEASE-SPECIFIC PAYLOADS

mRNA encoding for Chimeric Antigen Receptors (CARs), gene editing machinery, and other therapeutic proteins.

FIG. 1 | REDUCED LIVER DELIVERY WITH PREFERENTIAL BIODISTRIBUTION OF CAPSTAN tLNPS TO TARGET TISSUES

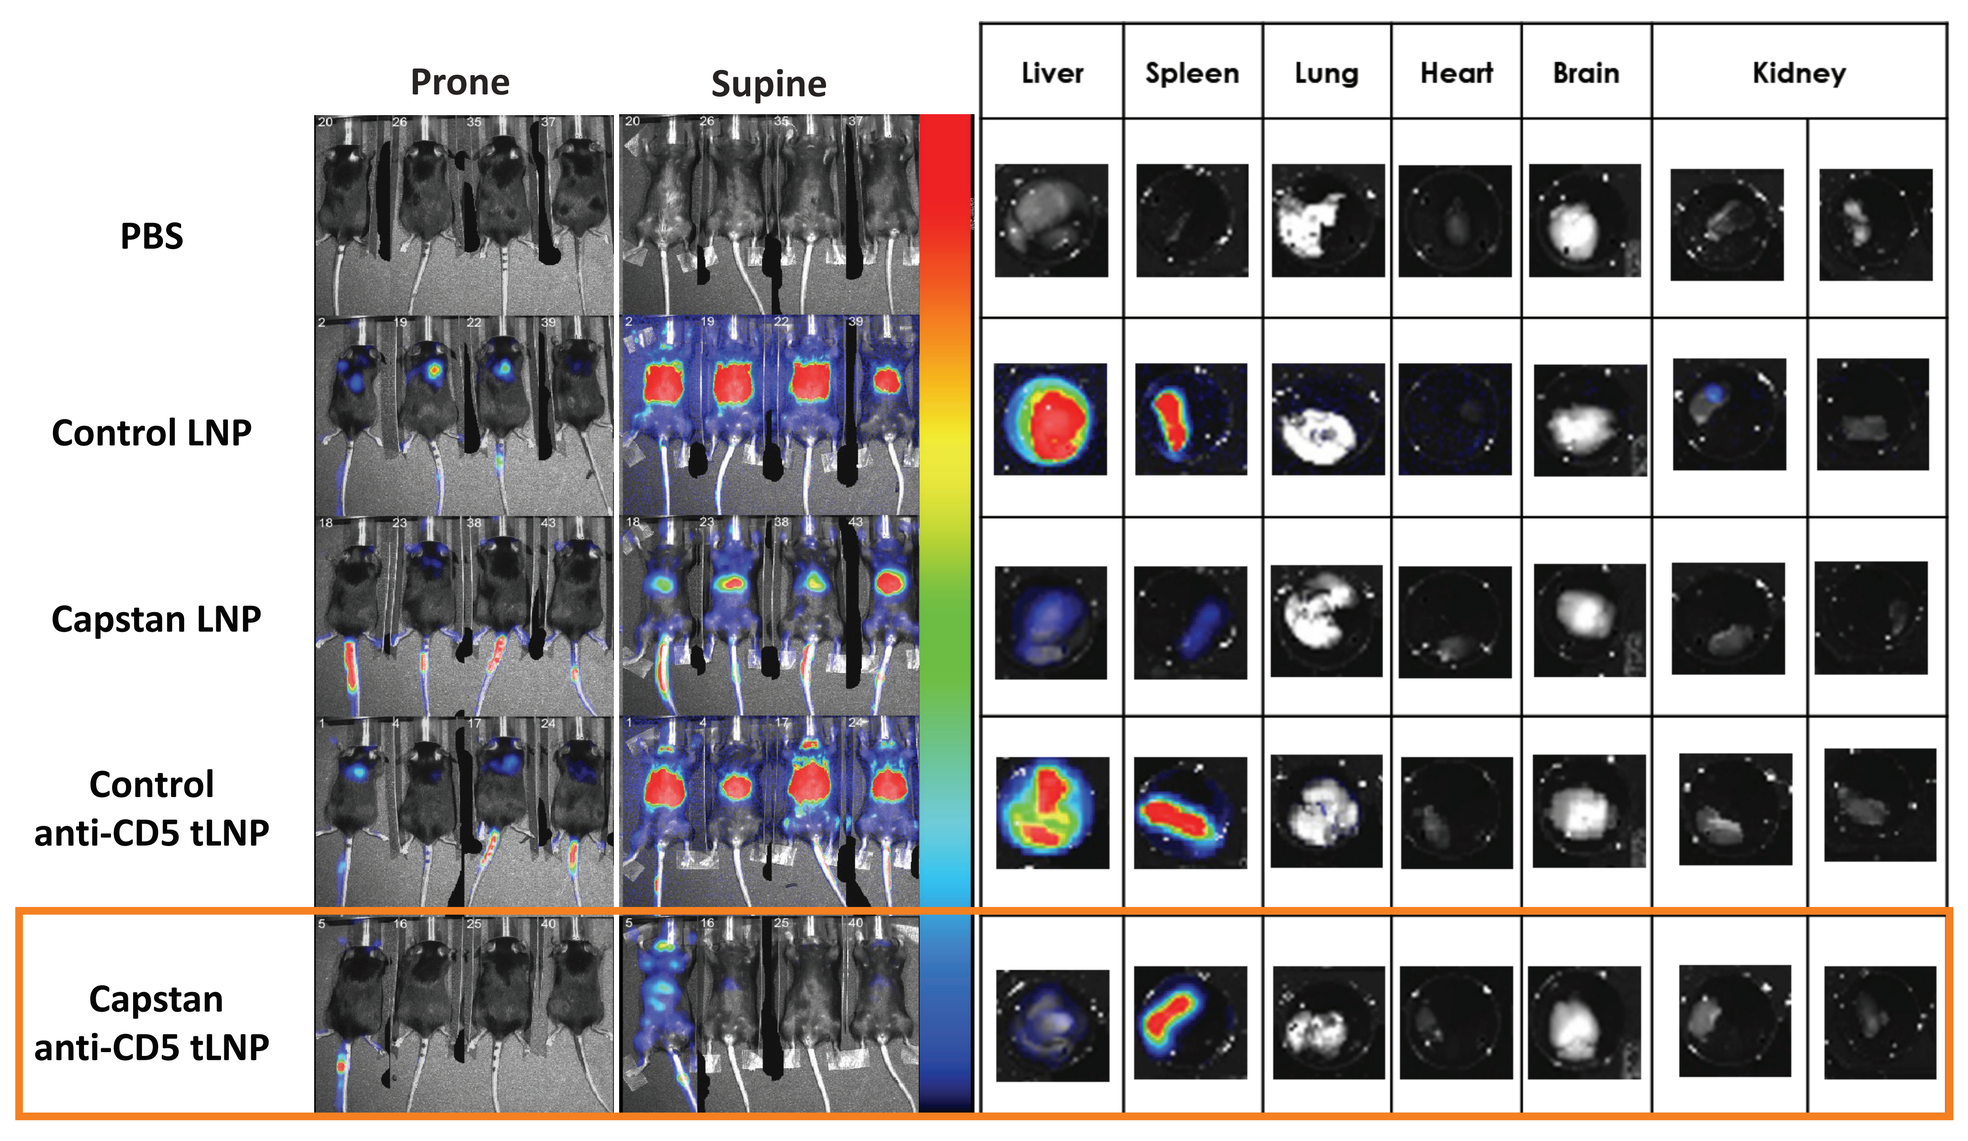
Prone
Supine
Liver
Spleen
Lung
Heart
Brain
Kidney
PBS
Control LNP
Capstan LNP
Control anti-CD5 tLNP
Capstan anti-CD5 tLNP

Payload: Luc mRNA

Targeting Ab: anti-mouse CD5

LNP: LNP without targeting Antibody

Control LNP: LNP designed for vaccines

Capstan LNP: Novel Ionizable lipid

tLNP: LNP conjugated to Targeting mAb

Nine-week-old female C57Bl/6 mice, (Charles River Laboratories) were intravenously injected with LNPs or tLNPs at a dose of 2 µg/animal via the tail vein. At 6 hours post-injection, prone and supine BLI images were collected from all mice. Following imaging, the mice were sacrificed, perfused and the following tissues collected: liver, spleen, lung, kidneys (both), heart and brain. Tissues were placed into luciferin pre-filled black polystyrene well plates with spacing between tissues and BLI images were collected.

FIG. 2 | CAPSTAN tLNPs DELIVER TO TARGET CELLS WITHIN TARGET TISSUES

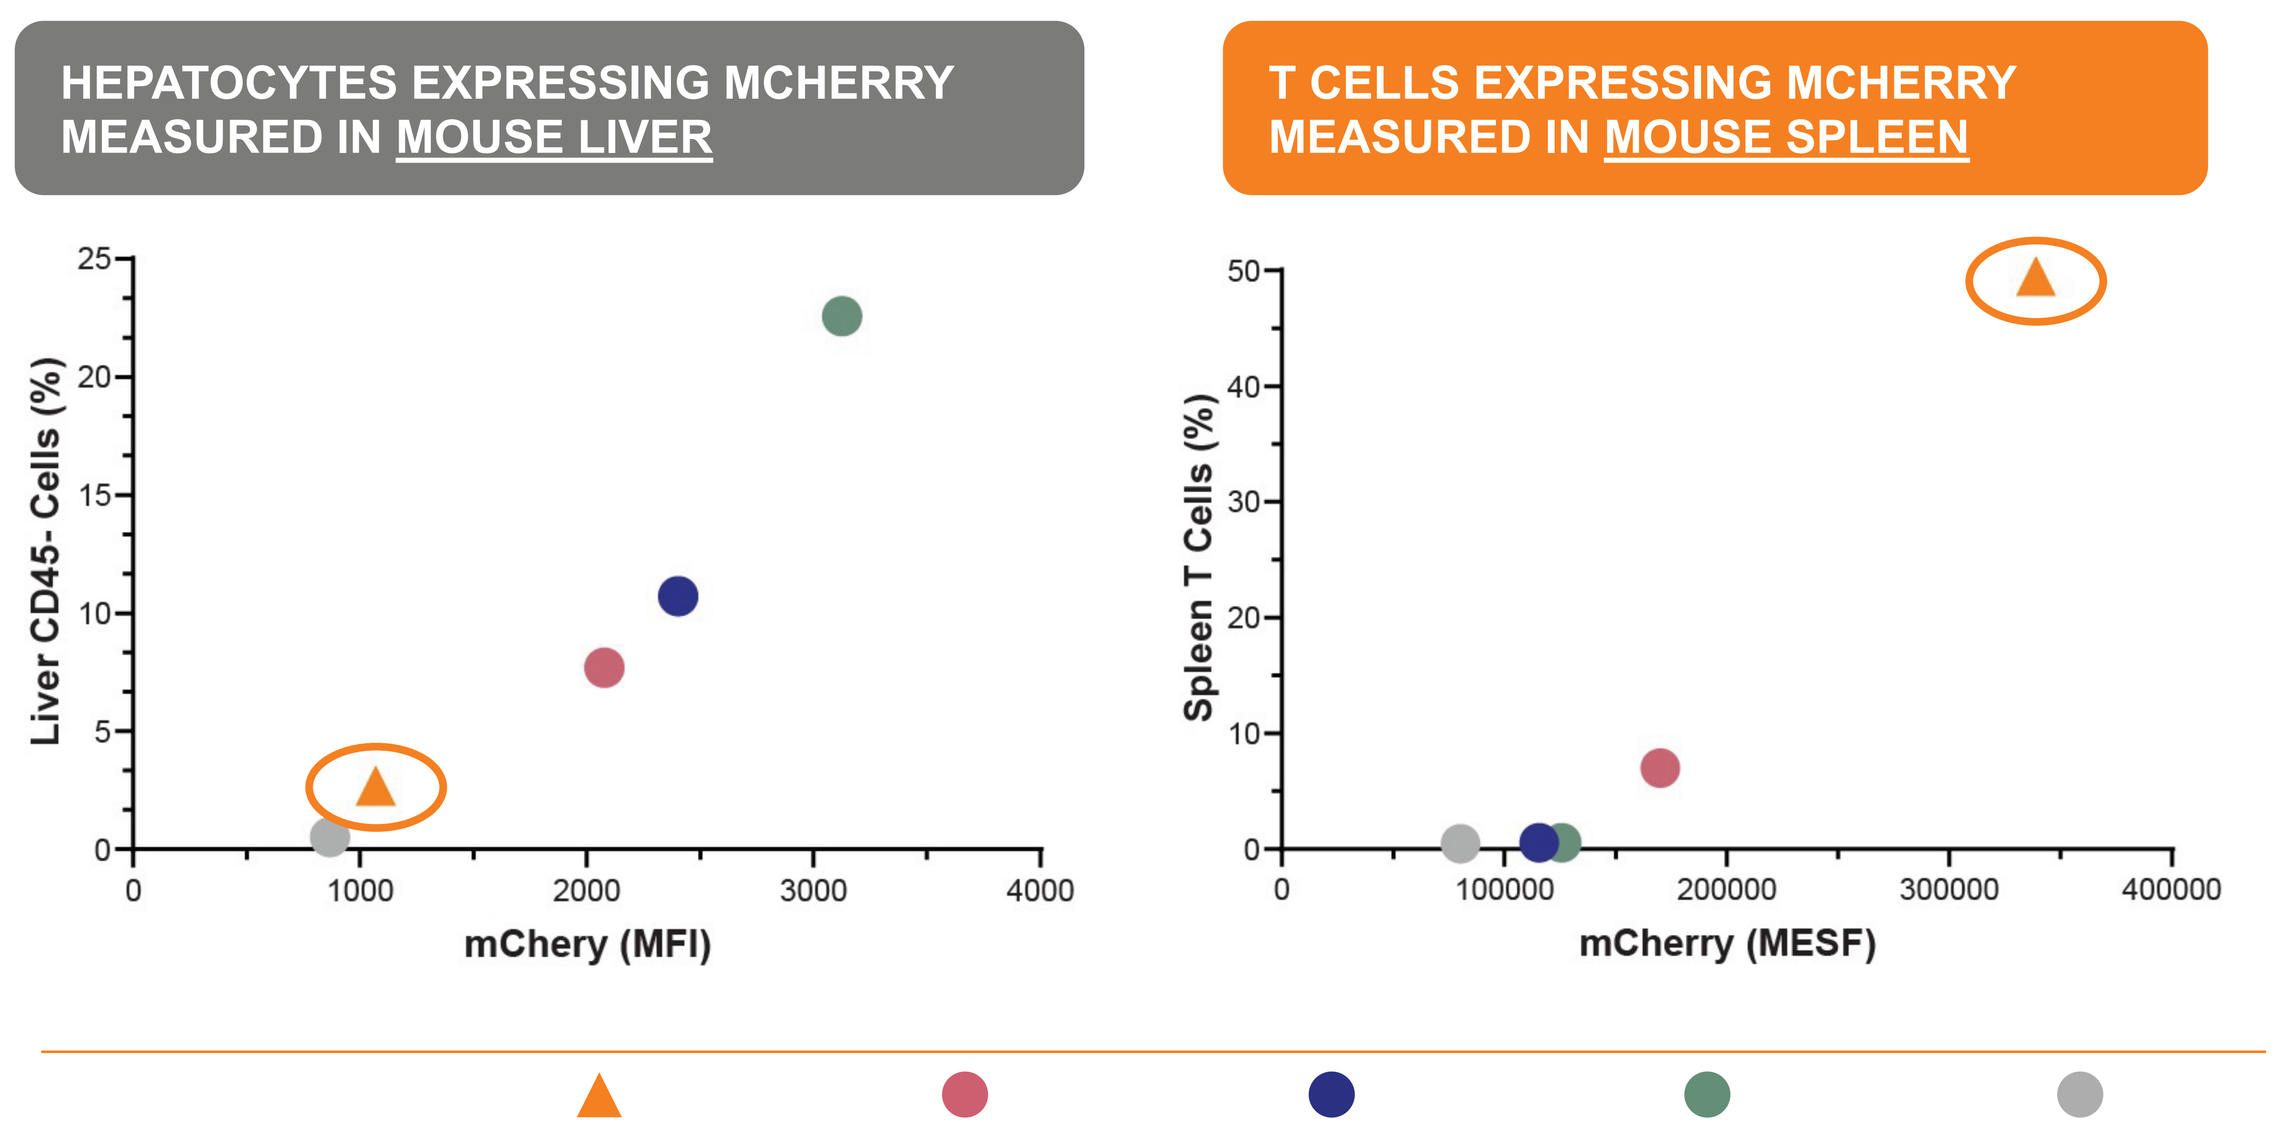
HEPATOCYTES EXPRESSING MCHERRY MEASURED IN MOUSE LIVER
T CELLS EXPRESSING MCHERRY MEASURED IN MOUSE SPLEEN
Liver CD45- Cells (%)
mChery (MFI)
Spleen T Cells (%)
mCherry (MESF)

	▲	●	●	●	●
Targeting mAb	CD5	CD5	Untargeted IgG	None	PBS
LNP	Capstan	Control	Control	Control	PBS

Payload: mCherry reporter mRNA
Targeting Ab: anti-mouse CD5

Control LNP: LNP designed for vaccines
MFI: Mean Fluorescence Intensity
MESF: Molecules of Equivalent Soluble Fluorochrome

8-10 week old female C57Bl/6 mice (Charles River Laboratories) were intravenously injected with LNPs or tLNPs at a dose of 10 µg/animal via the tail vein. At 24 hours post-injection, the mice were sacrificed, and the spleen and liver were collected. Tissues were disaggregated to single cell suspensions and analyzed for levels of mCherry expression by flow cytometry. The left panel depicts transfection rate (percentage of cells expressing mCherry) versus expression level (MFI) for hepatic CD45- cells. The right panel depicts transfection rate (percentage of cells expressing mCherry) versus expression level (MFI) for splenic CD3+ T-cells.

FIG. 3 | CAPSTAN tLNPs ARE WELL-TOLERATED IN RATS

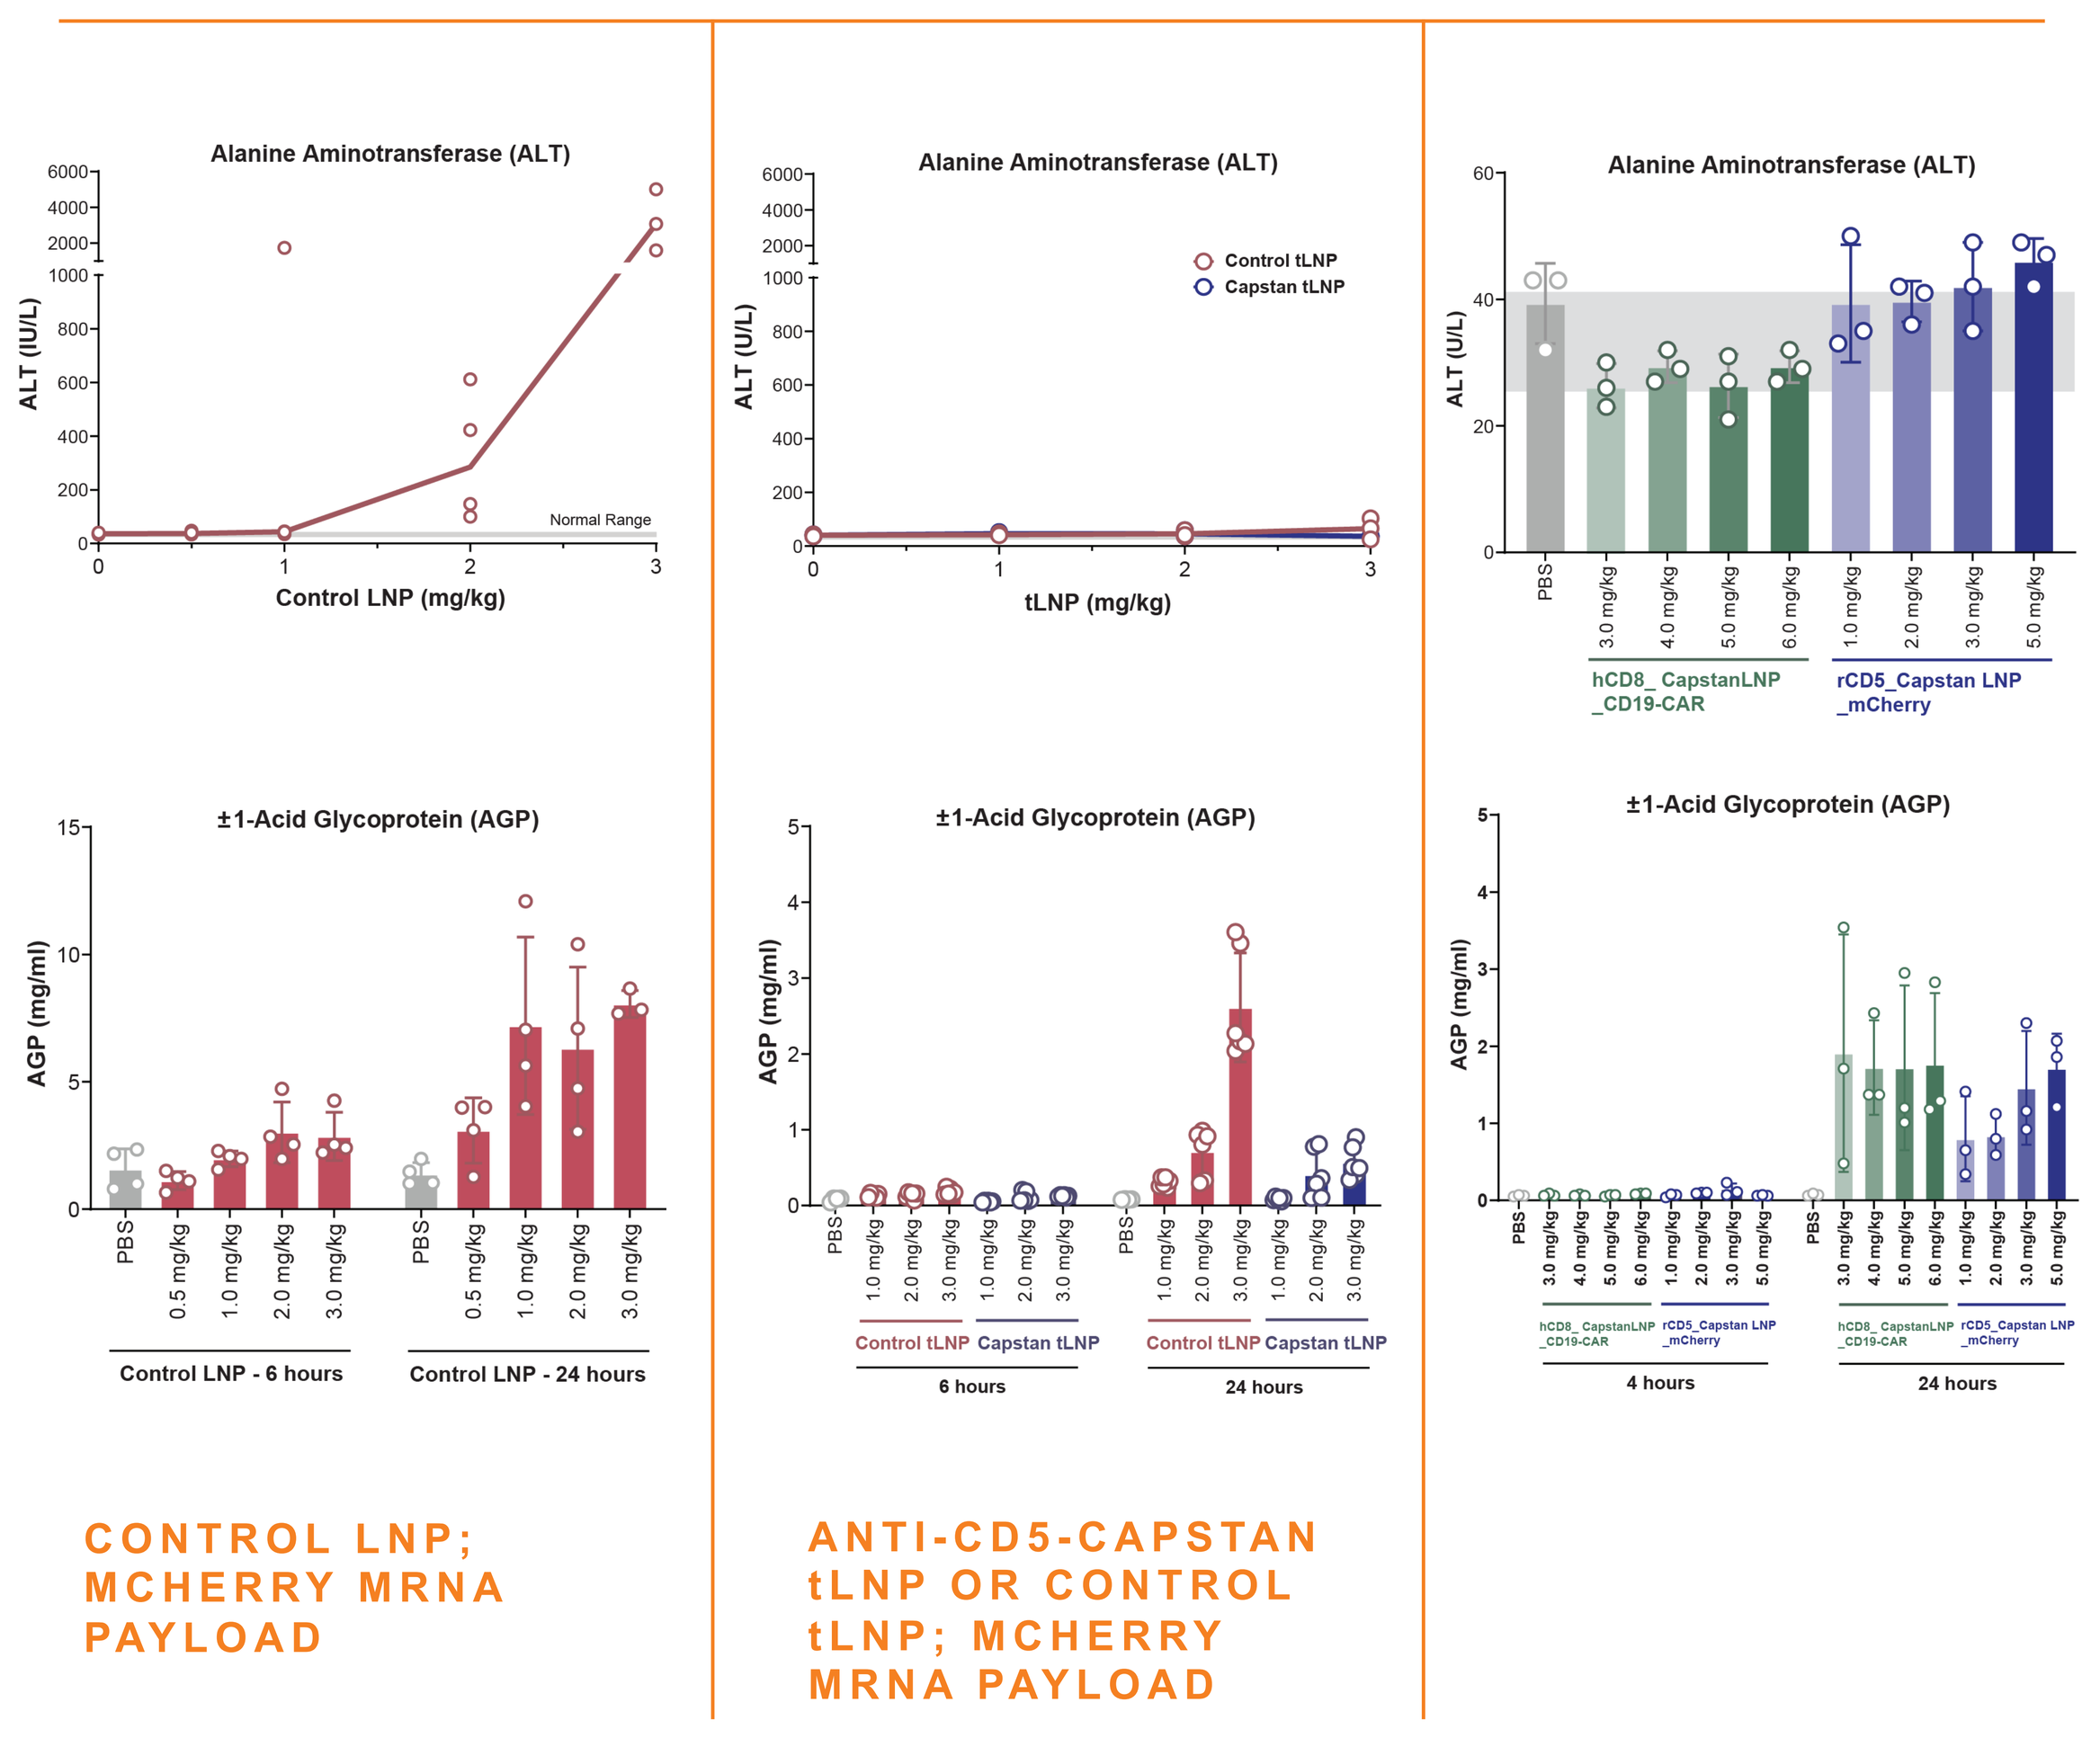
Alanine Aminotransferase (ALT)
Control LNP (mg/kg)
tLNP (mg/kg)
Control tLNP
Capstan tLNP
±1-Acid Glycoprotein (AGP)
Control LNP - 6 hours
Control LNP - 24 hours
hCD8_CapstanLNP_CD19-CAR
rCD5_Capstan LNP_mCherry
4 hours
24 hours
CONTROL LNP; MCHERRY MRNA PAYLOAD
ANTI-CD5-CAPSTAN tLNP OR CONTROL tLNP; MCHERRY MRNA PAYLOAD

Levels of an acute phase protein and a liver enzyme in rats administered the control LNP and tLNP or Capstan LNP or tLNP. Specifically, α1-acid glycoprotein levels were measured by Luminex at 6 and 24 hours post-administration of the LNPs or tLNPs. Alanine aminotransferase levels were measured at 24 hours post-treatment. Control LNPs contain a widely utilized ionizable lipid used in vaccines.

FIG. 4 | ANTIBODY-BASED LNP TARGETING ENABLES SELECTIVE ENGINEERING OF HUMAN TARGET CELLS

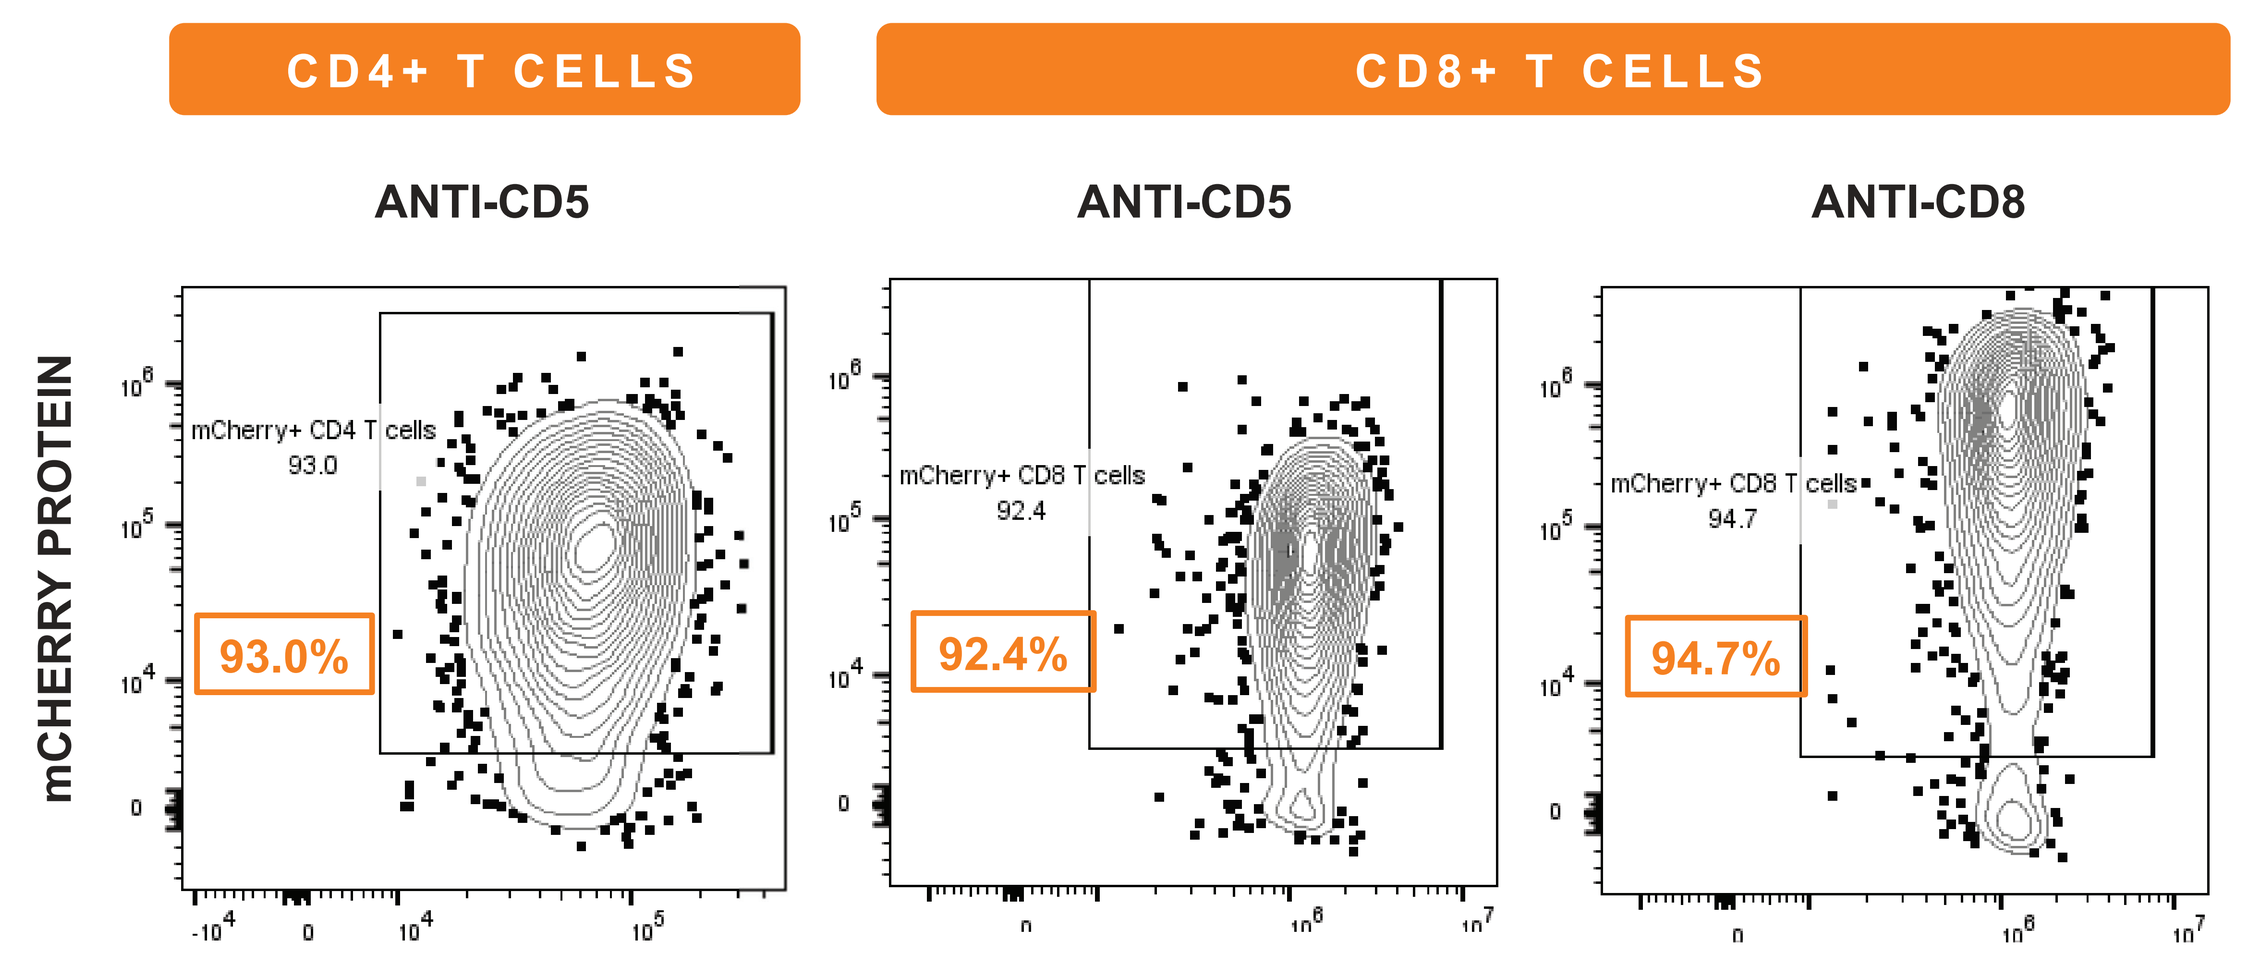
CD4+ T CELLS
CD8+ T CELLS
ANTI-CD5
ANTI-CD5
ANTI-CD8
mCHERRY PROTEIN
93.0%
92.4%
94.7%

NSG mice (approximately 10 weeks old) were purchased from The Jackson Laboratory and acclimated for at least 5 days. 10 million human peripheral blood mononuclear cells (PBMCs) were injected intravenously via the tail vein. After 17 days of engraftment mice were evaluated for frequency of human CD45+ cells in circulation and staged in groups with similar averages. Groups of mice were injected intravenously with a single dose (10 µg/animal) of human CD5-targeting tLNPs with mCherry or human CD8-targeting tLNPs with mCherry mRNA. Mice were sacrificed 24 hours after dosing to assess mCherry expression in T-cell subsets in the spleen.

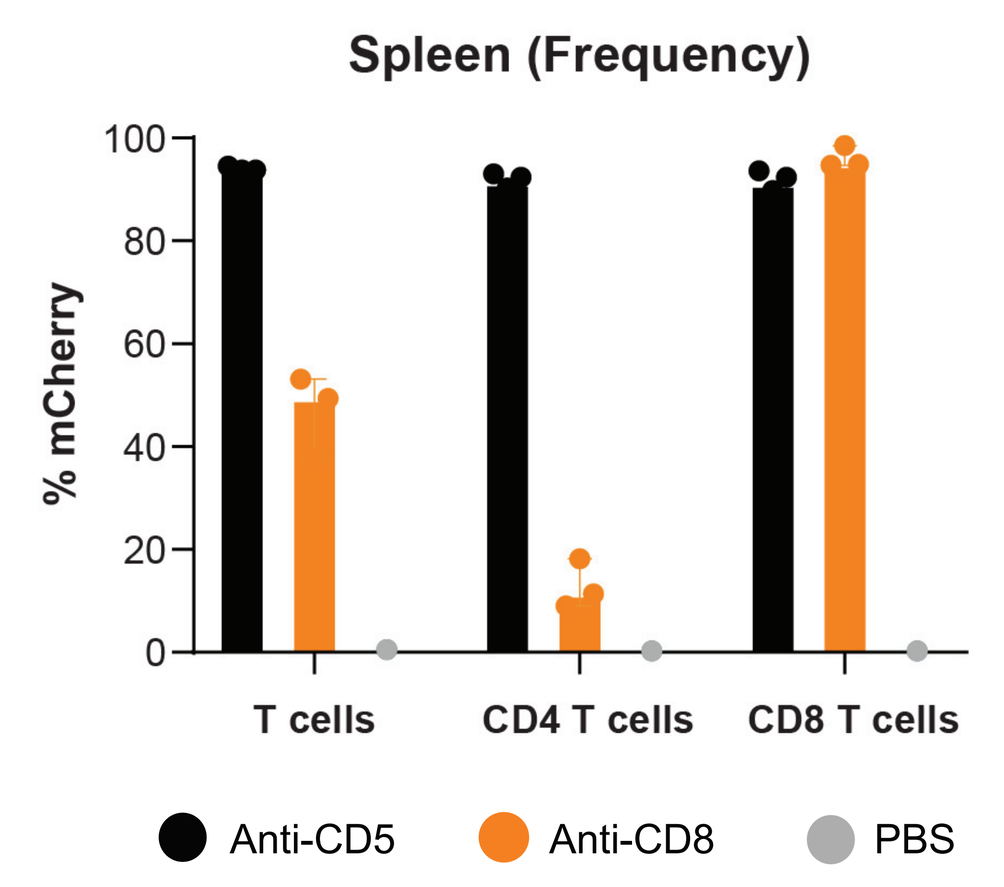
Spleen (Frequency)
% mCherry
T cells
CD4 T cells
CD8 T cells
Anti-CD5
Anti-CD8
PBS

FIG. 5 | CAR AND mRNA SEQUENCE OPTIMIZATIONS RESULT IN INCREASED EXPRESSION AND FUNCTION

***IN VITRO* CAR FUNCTION: NALM6 KILLING BY FRESHLY ACTIVATED HUMAN T CELLS**

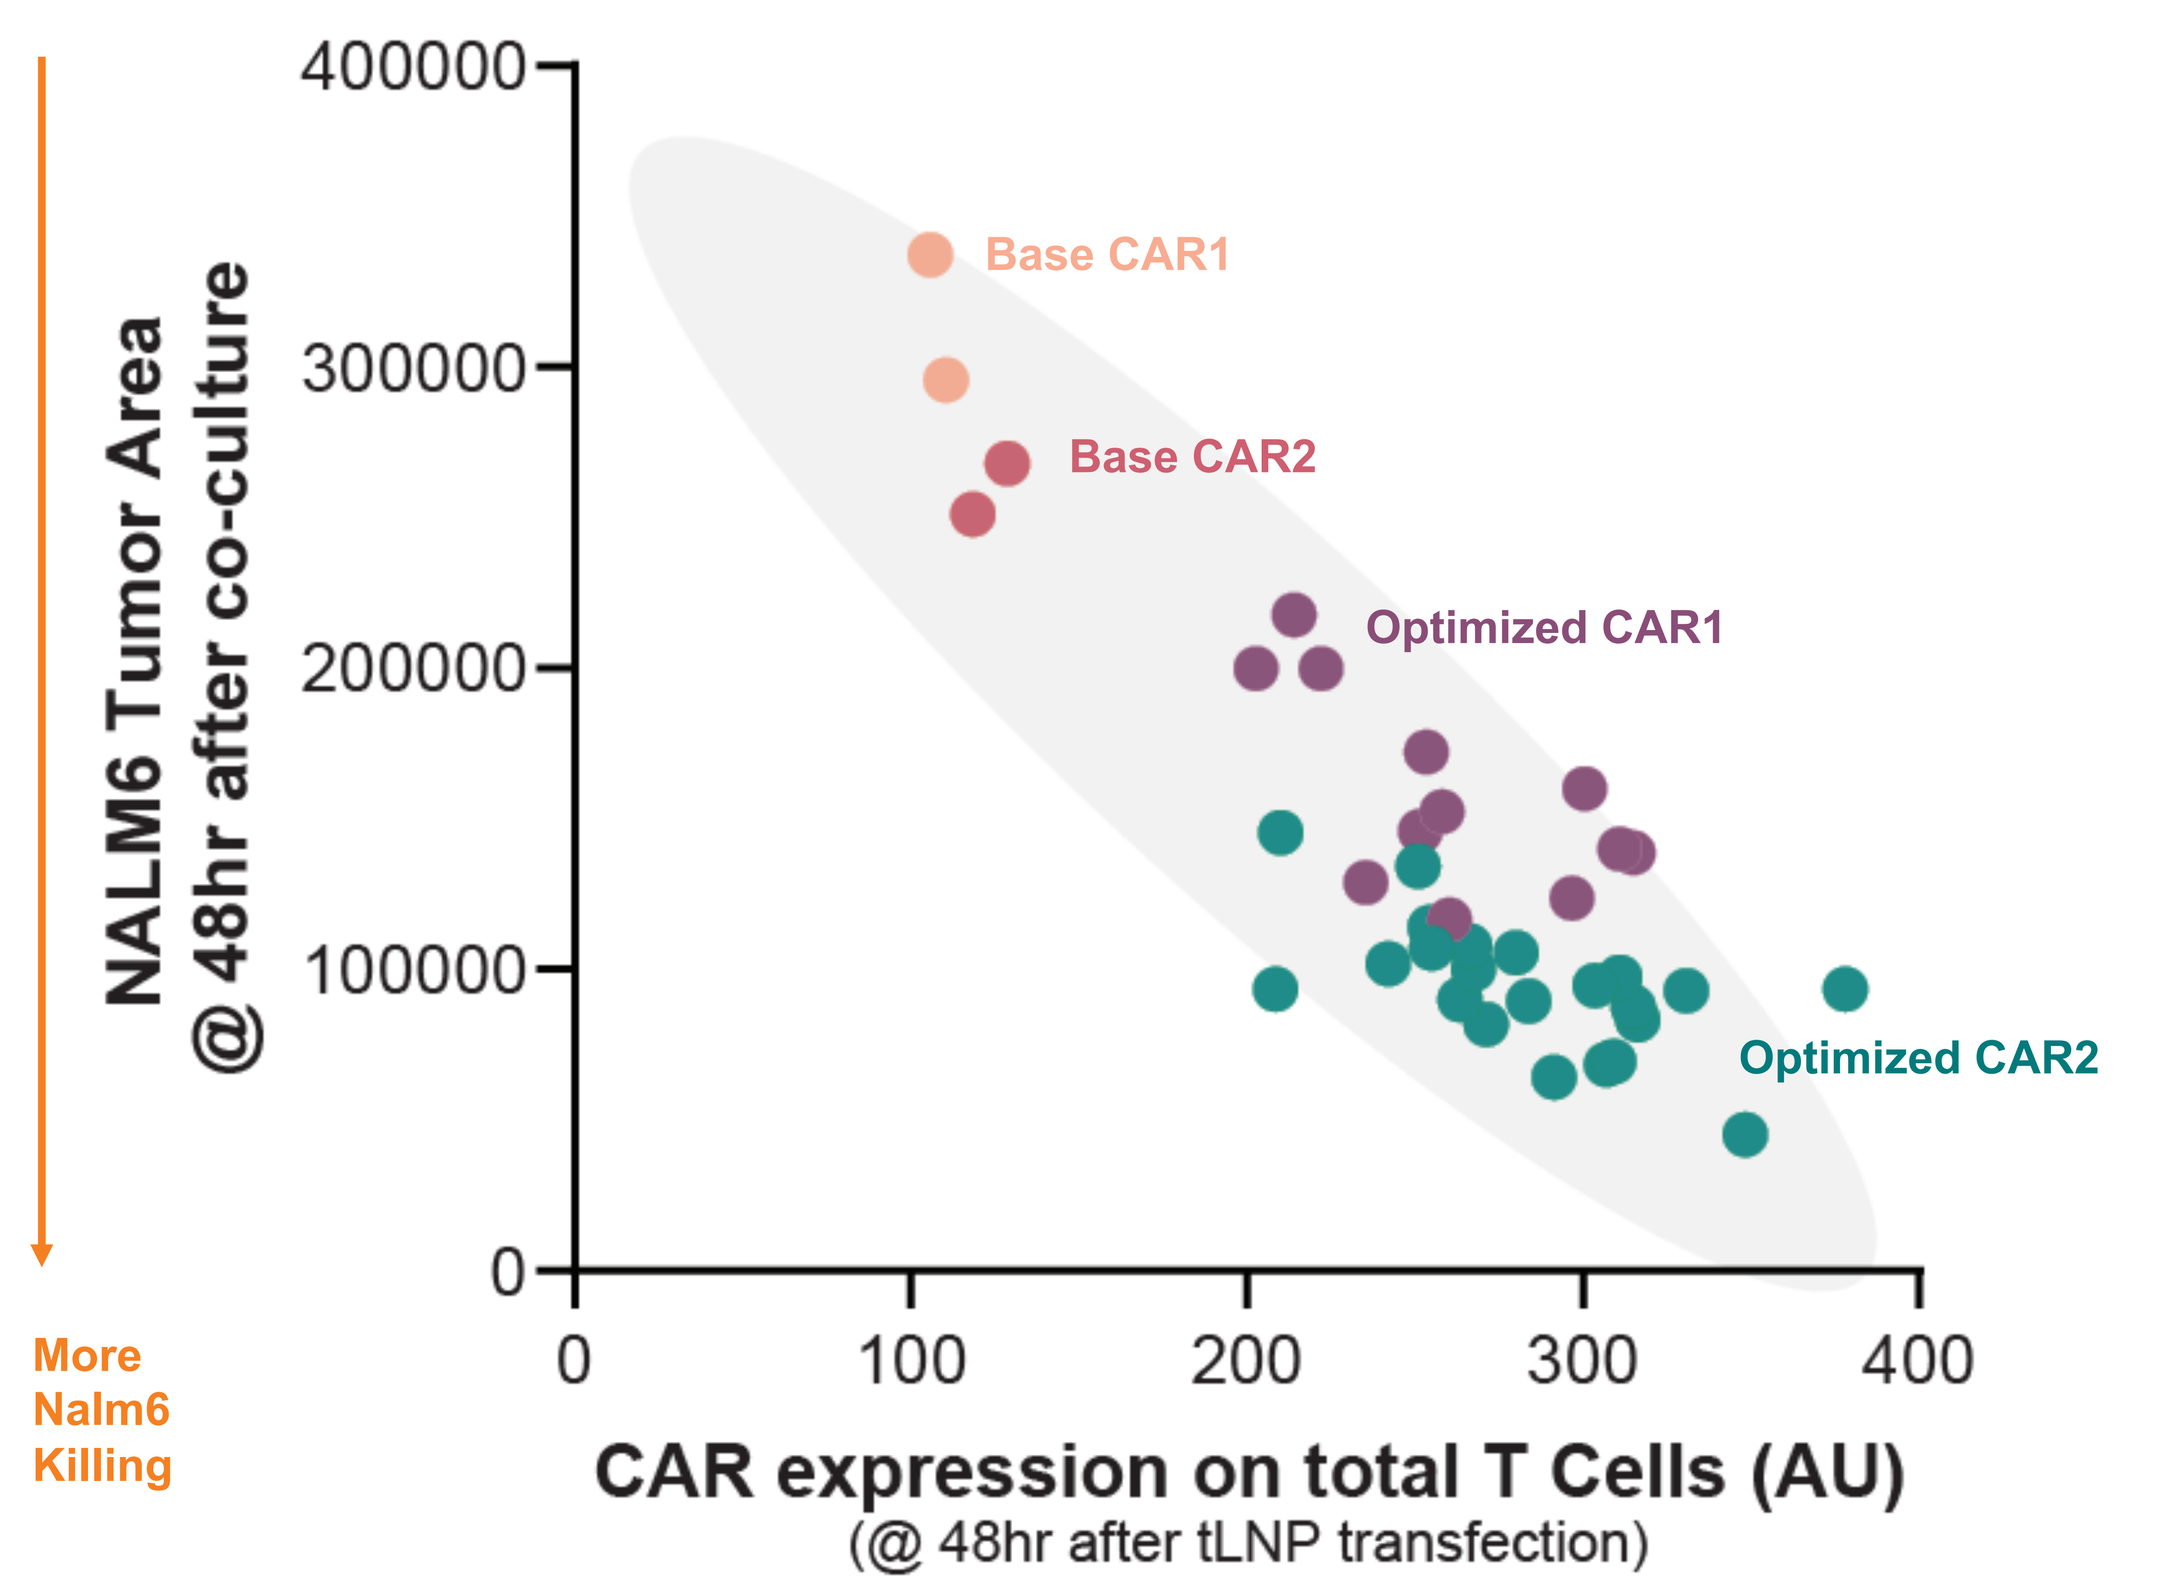
NALM6 Tumor Area @ 48hr after co-culture
More Nalm6 Killing
Base CAR1
Base CAR2
Optimized CAR1
Optimized CAR2
CAR expression on total T Cells (AU)
(@ 48hr after tLNP transfection)

The graph shows CD19 CAR molecules per cell on total T-cells from CAR1 encoding mRNA and CAR2 encoding mRNA 48-hours post tLNP transfection and GFP signals from Nalm6 cells after 48-hours of co-culture. The data points from base constructs are also shown. CAR1 and CAR2 share the same CD19 binding moiety.

FIG. 6 | CAPSTAN tLNP TREATMENT RESULTS IN RAPID AND DEEP PRIMARY HUMAN B CELL DEPLETION IN HUMANIZED MICE

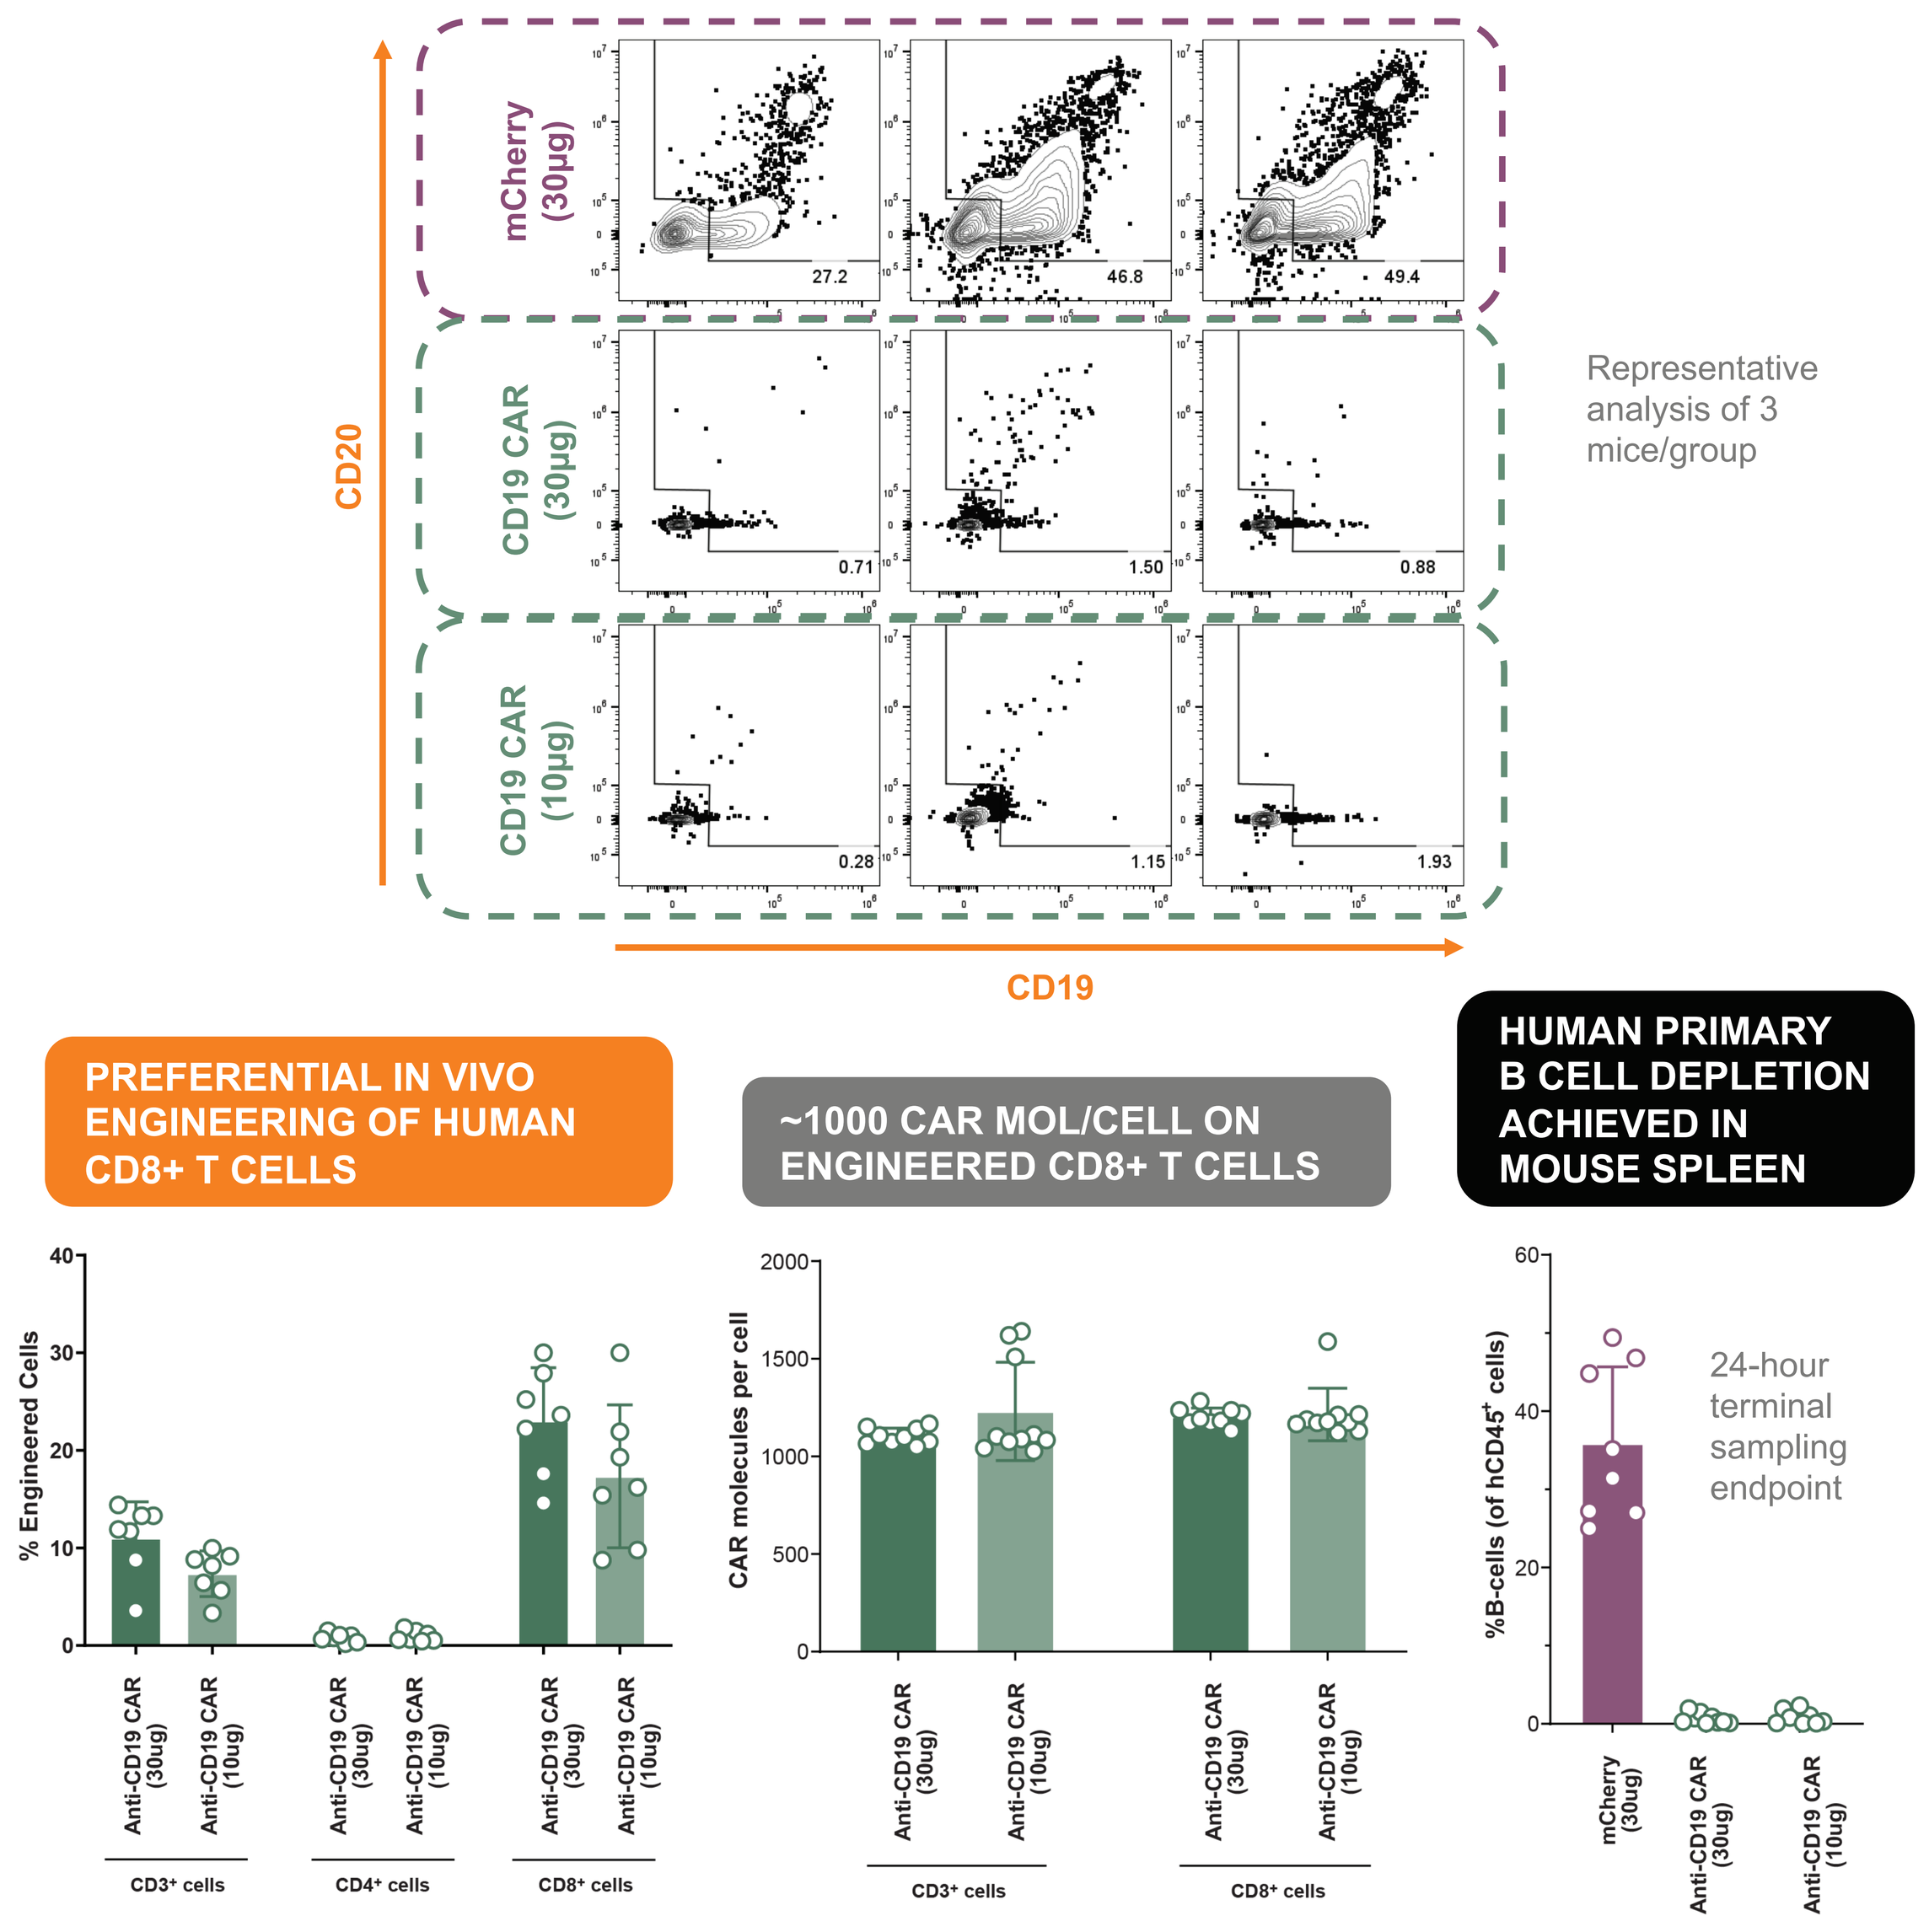
mCherry (30µg)
CD19 CAR (30µg)
CD19 CAR (10µg)
CD20
CD19
Representative analysis of 3 mice/group
PREFERENTIAL IN VIVO ENGINEERING OF HUMAN CD8+ T CELLS
~1000 CAR MOL/CELL ON ENGINEERED CD8+ T CELLS
HUMAN PRIMARY B CELL DEPLETION ACHIEVED IN MOUSE SPLEEN
% Engineered Cells
CAR molecules per cell
%B-cells (of hCD45+ cells)
24-hour terminal sampling endpoint

NSG mice (approximately 10 weeks old) were purchased from The Jackson Laboratory and acclimated for at least 5 days. Ten million human peripheral blood mononuclear cells (PBMCs) were injected intravenously via the tail vein. After 17 days of engraftment mice were evaluated for frequency of human CD45+ cells in circulation and staged in groups with similar averages. Groups of mice were injected intravenously with a single dose (either 30 µg/animal or 10 µg/animal) of human CD8-targeting tLNPs with an anti-CD19 CAR mRNA, or 30 µg/animal of a human CD8-targeting tLNPs with mCherry. Mice were sacrificed 24 hours after dosing to assess B cell levels.

FIG. 7 | RAPID TUMOR CONTROL IN HUMANIZED MICE WITH tLNPs TARGETING PAN T CELLS AND T CELL SUBSETS

A. BIOLUMINESCENCE IMAGING OF NALM6 TUMORS

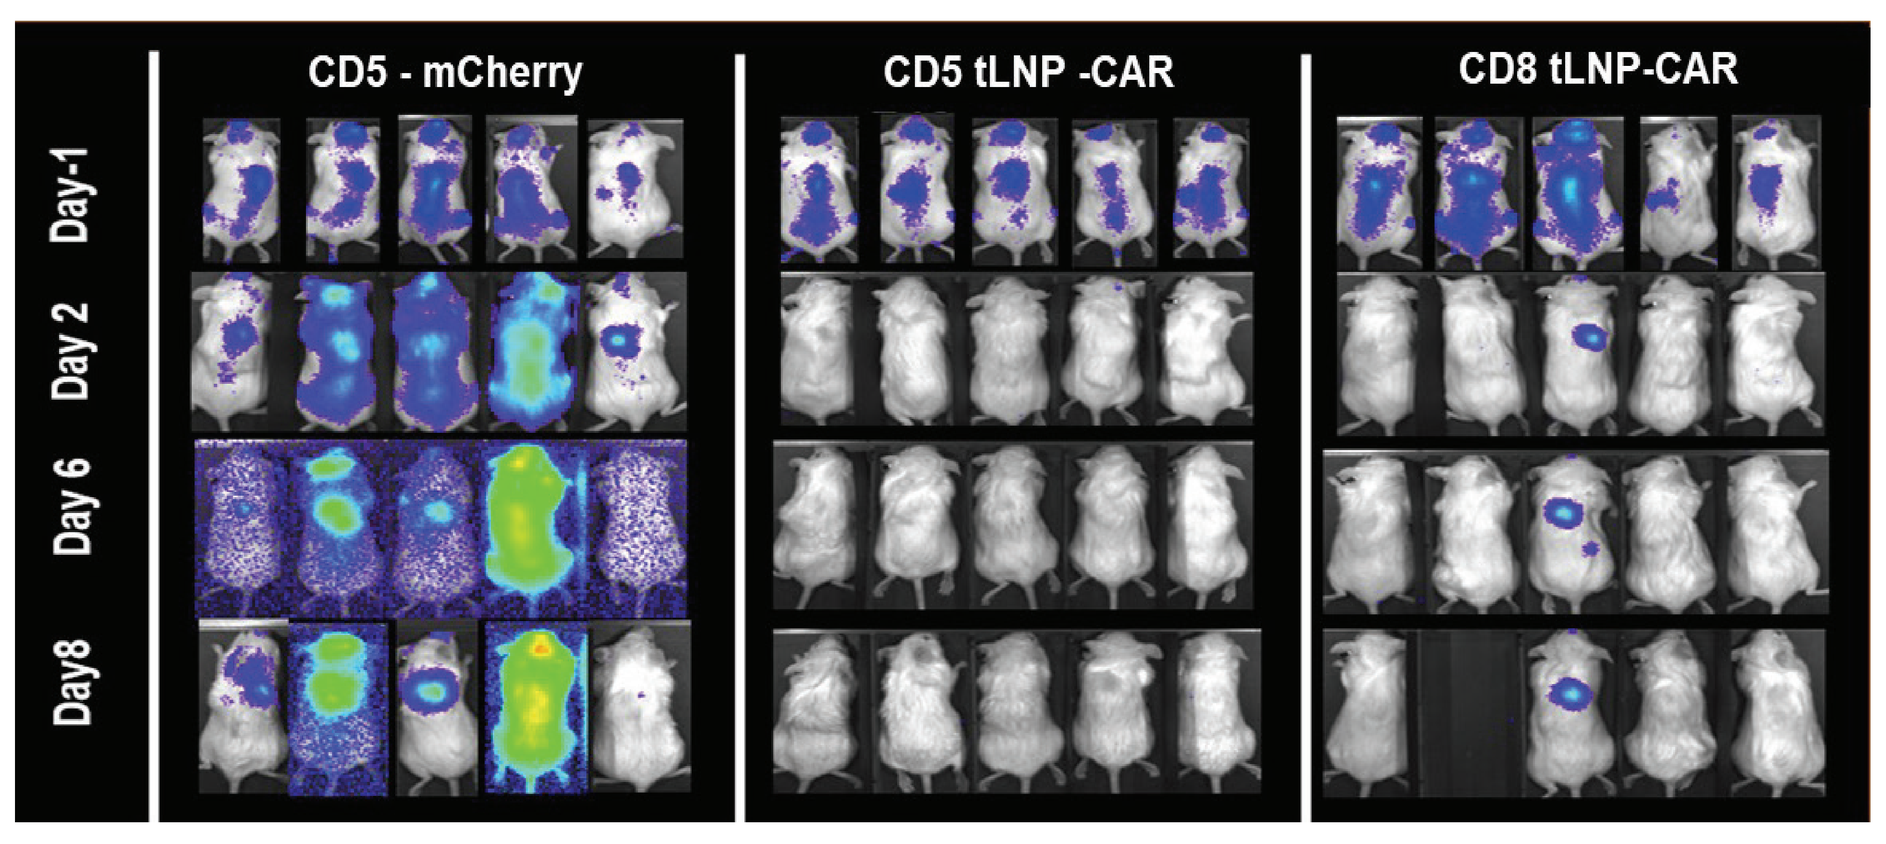
CD5 - mCherry
CD5 tLNP -CAR
CD8 tLNP-CAR
Day-1
Day 2
Day 6
Day8

Both treatments control tumor growth as early as 2 days post first treatment after only 1 dose of tLNP targeting either CD5 or CD8

B. TUMOR MEASUREMENTS

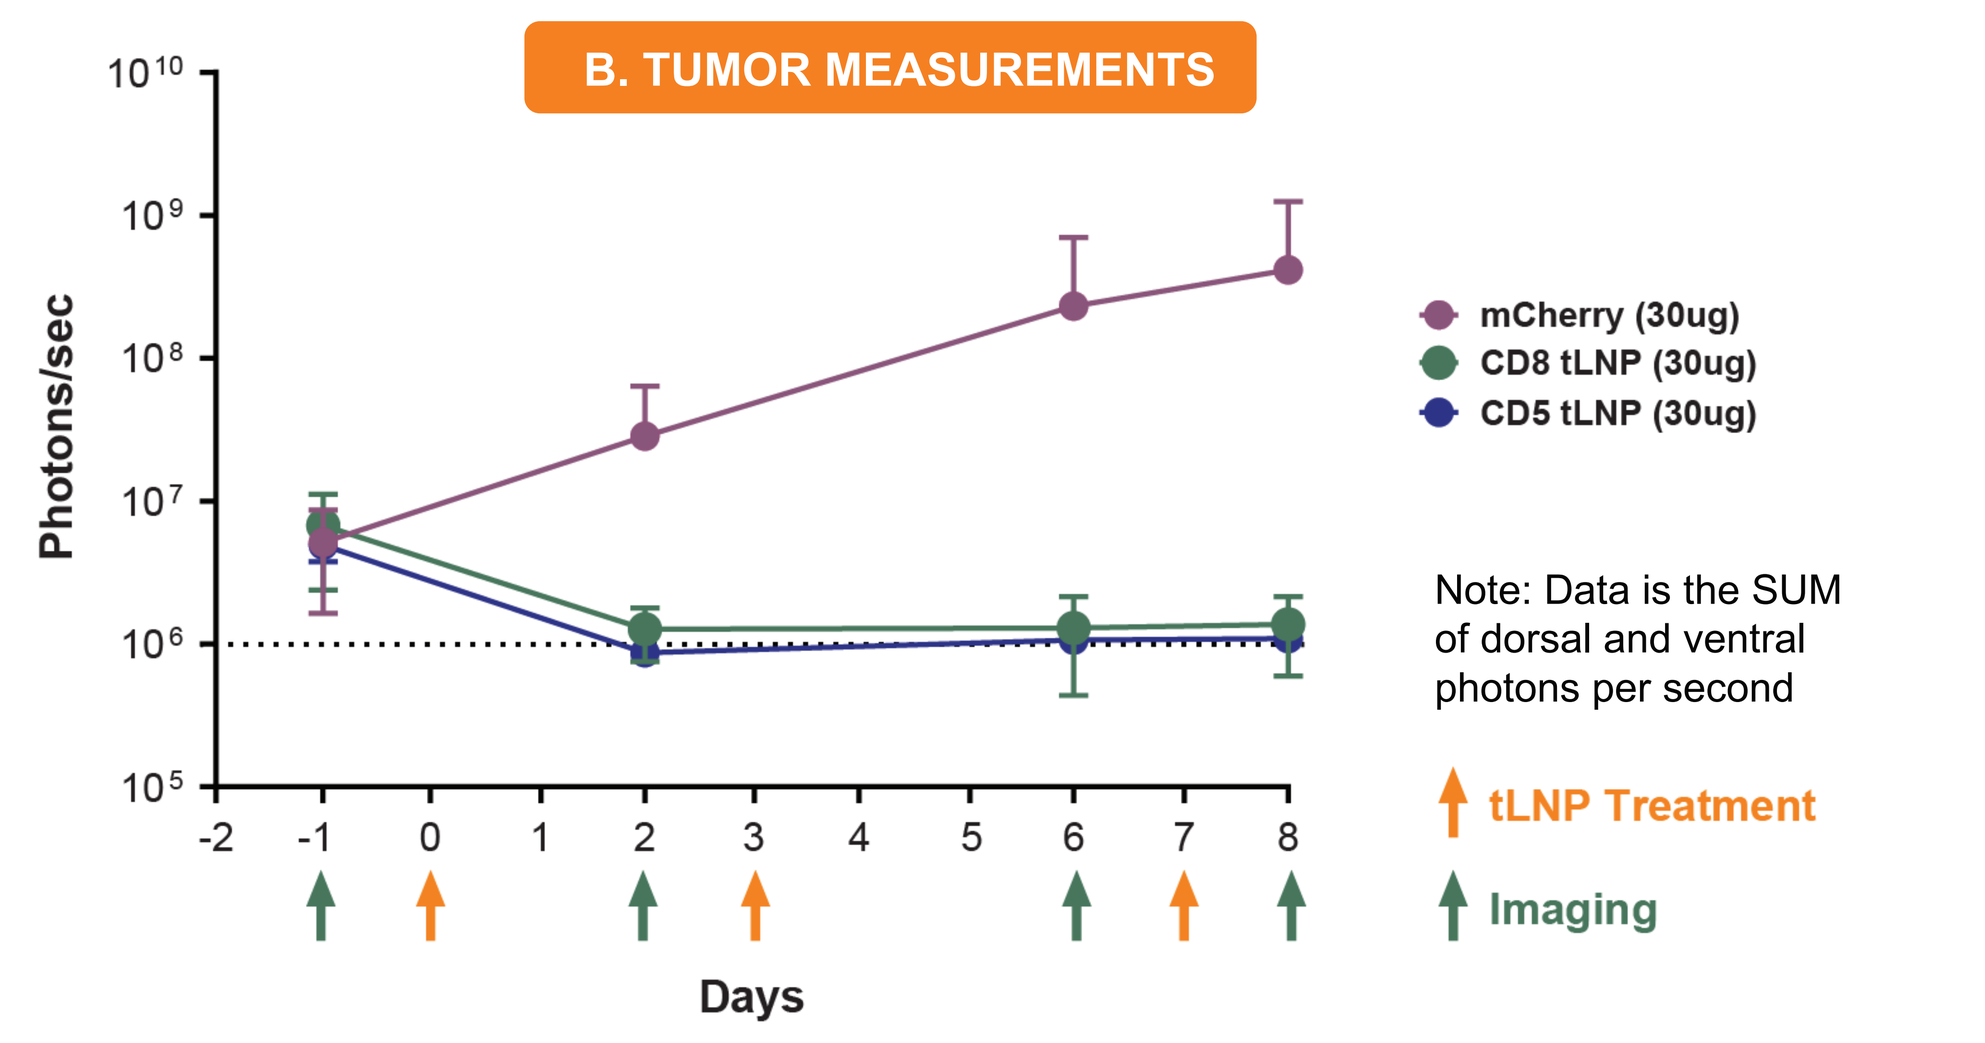
Photons/sec
Days
mCherry (30ug)
CD8 tLNP (30ug)
CD5 tLNP (30ug)
Note: Data is the SUM of dorsal and ventral photons per second
tLNP Treatment
Imaging

C. CAR T CELL EXPRESSION IN VIVO

24 hrs post 3rd tLNP infusion

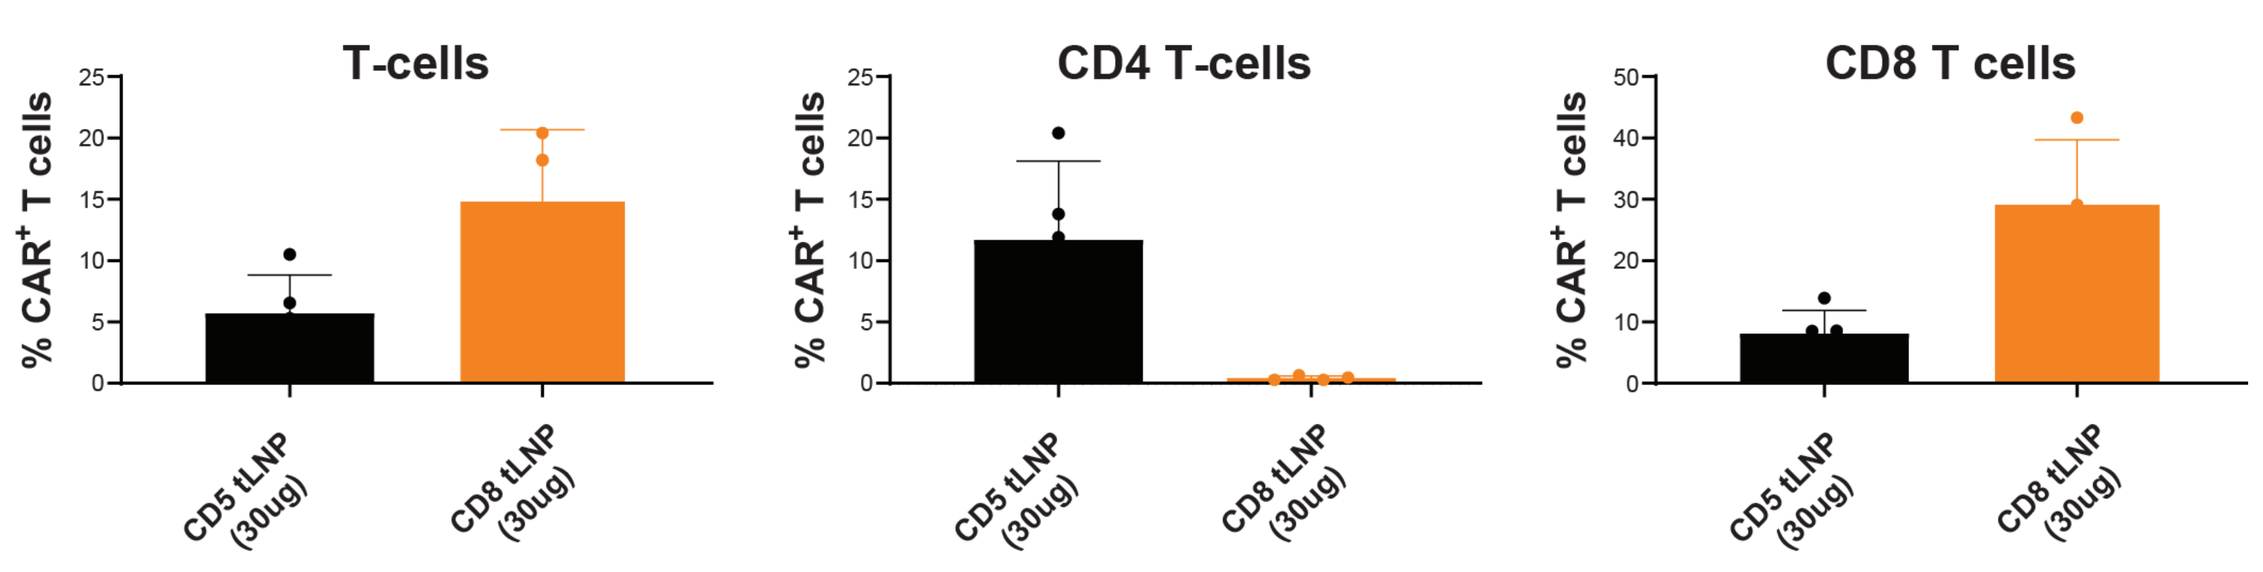
T-cells
CD4 T-cells
CD8 T cells
% CAR+ T cells
CD5 tLNP (30ug)
CD8 tLNP (30ug)

NSG mice (approximately 10 weeks old) were purchased from The Jackson Laboratory and acclimated for at least 5 days. Ten million human T cells were injected intravenously via the tail vein. After 10 days of the T cell engraftment, 5.0E+05 Nalm6 cells constitutively expressing Firefly Luciferase (Nalm6-Luc) were injected intravenously. Seven days following tumor cell engraftment mice were evaluated for T cell engraftment (frequency of human CD45+ cells in circulation) and tumor burden (assessed by Luciferase signal) and staged in groups with similar averages of both readouts. From the 18th day following T cell engraftment, groups of mice were injected intravenously with human CD5-targeting tLNPs with an anti-CD19 CAR mRNA, human CD8-targeting tLNPs with an anti-CD19 CAR mRNA, or human CD5-targeting tLNPs with mCherry mRNA twice weekly for a total of 5 doses. Tumor cell burden was evaluated by bioluminescence imaging (BLI) of the luciferase signal twice weekly. 24 hrs after the third dose, peripheral blood samples were analyzed for CAR expression on T cells.

CONCLUSIONS

- Capstan has developed rationally designed proprietary LNPs that effectively target and engineer T cells in vivo when functionalized with a T cell specific antibody and exhibit significantly reduced delivery to liver.
- These tLNPs were well tolerated following a single intravenous dose up through 6 mg/kg in male rats
- Capstan's tLNPs can specifically target and deliver a therapeutic CAR payload to human T cells in vivo resulting in functional anti-tumor or anti-primary B cell activity.
- Treatments for various disease categories using tLNPs can be envisioned through different targeting binders to deliver a broad set of payloads to diverse cell populations.